Biomarkers of SQ House Dust Mite Sublingual Immunotherapy (SLIT)-Tablet Treatment After Nasal Allergen Challenge

Gunawardana, N.C.[1], Zhao, Q.[2], Tsai, K.[2], Selverian, D.[2], Carayannopoulos, L.N.[2], Lund, K.[3], Clarke, G.W.[4]*, Mant, T.[4], Hansel, T.T.[1], Nolte, H.[2]

[1]Imperial Clinical Respiratory Research Unit (ICRRU), St. Mary's Hospital, London, UK; [2]Merck & Co., Inc., Kenilworth, NJ, USA; [3]ALK, Hørsholm, Denmark; [4]QuintilesIMS, Reading, UK

Introduction

- Treatment with SQ house dust mite (HDM) sublingual immunotherapy (SLIT) tablet:
 - Improves allergic rhinoconjunctivitis symptoms
 - Improves time to first moderate or severe asthma exacerbation during inhaled corticosteroid reduction
 - Increases antibodies with the capacity to compete with IgE (including allergen-specific IgG_4 and functional blocking capacity measured as IgE blocking factor [BF])[1,2]
- There have been difficulties in establishing accurate, reliable biomarkers to assess allergy immunotherapy treatment response and little work surrounding the pharmacodynamics and biomarkers for HDM immunotherapy
- Nasal allergen challenge (NAC) allows the delivery of a controlled allergen stimulus to the nasal mucosa to evaluate the effects of immunotherapy on clinical symptoms and local nasal immune responses

Methods

Study design

- This was an exploratory, phase 1b, randomized, placebo-controlled, double-blind study (NCT01852825) conducted across two sites
- Adults with a clinical history of allergic rhinitis to HDM with demonstrable sensitization were randomized in a 2:1 ratio to receive daily 12 SQ-HDM (MK-8237, Merck & Co., Inc., Kenilworth, NJ, USA/ALK, Hørsholm, Denmark) or placebo for 12 weeks
- NAC was conducted at 2 weeks pre-treatment, week 8, and week 12 (**Figure 1**)
 - Nasal Pfeiffer Bidose applicator (Aptar Pharma, Milton Keynes, UK) was used to administer a challenge of 900 SQ-U HDM allergen extract (Aquagen, *D pteronyssinus*; ALK, Hørsholm, Denmark) per nostril
- Total nasal symptom score (TNSS) was assessed by a visual analog scale (VAS) from 0 (absent) to 100 (severe) for each symptom of nasal congestion, rhinorrhea, itching, and sneezing (total possible 400)
- TNSS and peak nasal inspiratory flow (PNIF) were recorded before NAC, every 15 minutes in the first hour, at 90 minutes, and hourly for 6 hours
- Nasal mucosal lining fluid (MLF) was collected using synthetic absorptive membranes by nasosorption, and concentrations of IL-5, IL-13, and TARC in MLF determined using singleplex immunoassays
- Nasal scrapes were collected using nasal curettes from the inferior side of the inferior turbinate at 1 hour pre-NAC and 6.5 hours post-NAC, and nasal mRNA assessed
- Serum for measurement of IgE-BF and IgG_4 were analyzed using validated immunoassays

Endpoints

- Primary endpoints were the change induced by 12 SQ-HDM from baseline in HDM-specific IgG_4 and IgE-BF antibodies at week 12
- Secondary endpoints were changes in nasal MLF IL-5 concentration in response to NAC after treatment, and changes in time-weighted average TNSS during NAC for early phase (baseline to 1 hour post NAC) and peak (15 minutes post NAC) responses

Methods (continued)

Figure 1. A) Study design; B) sampling schedule at weeks −2, 8, and 12. NAC, nasal allergen challenge.

A)

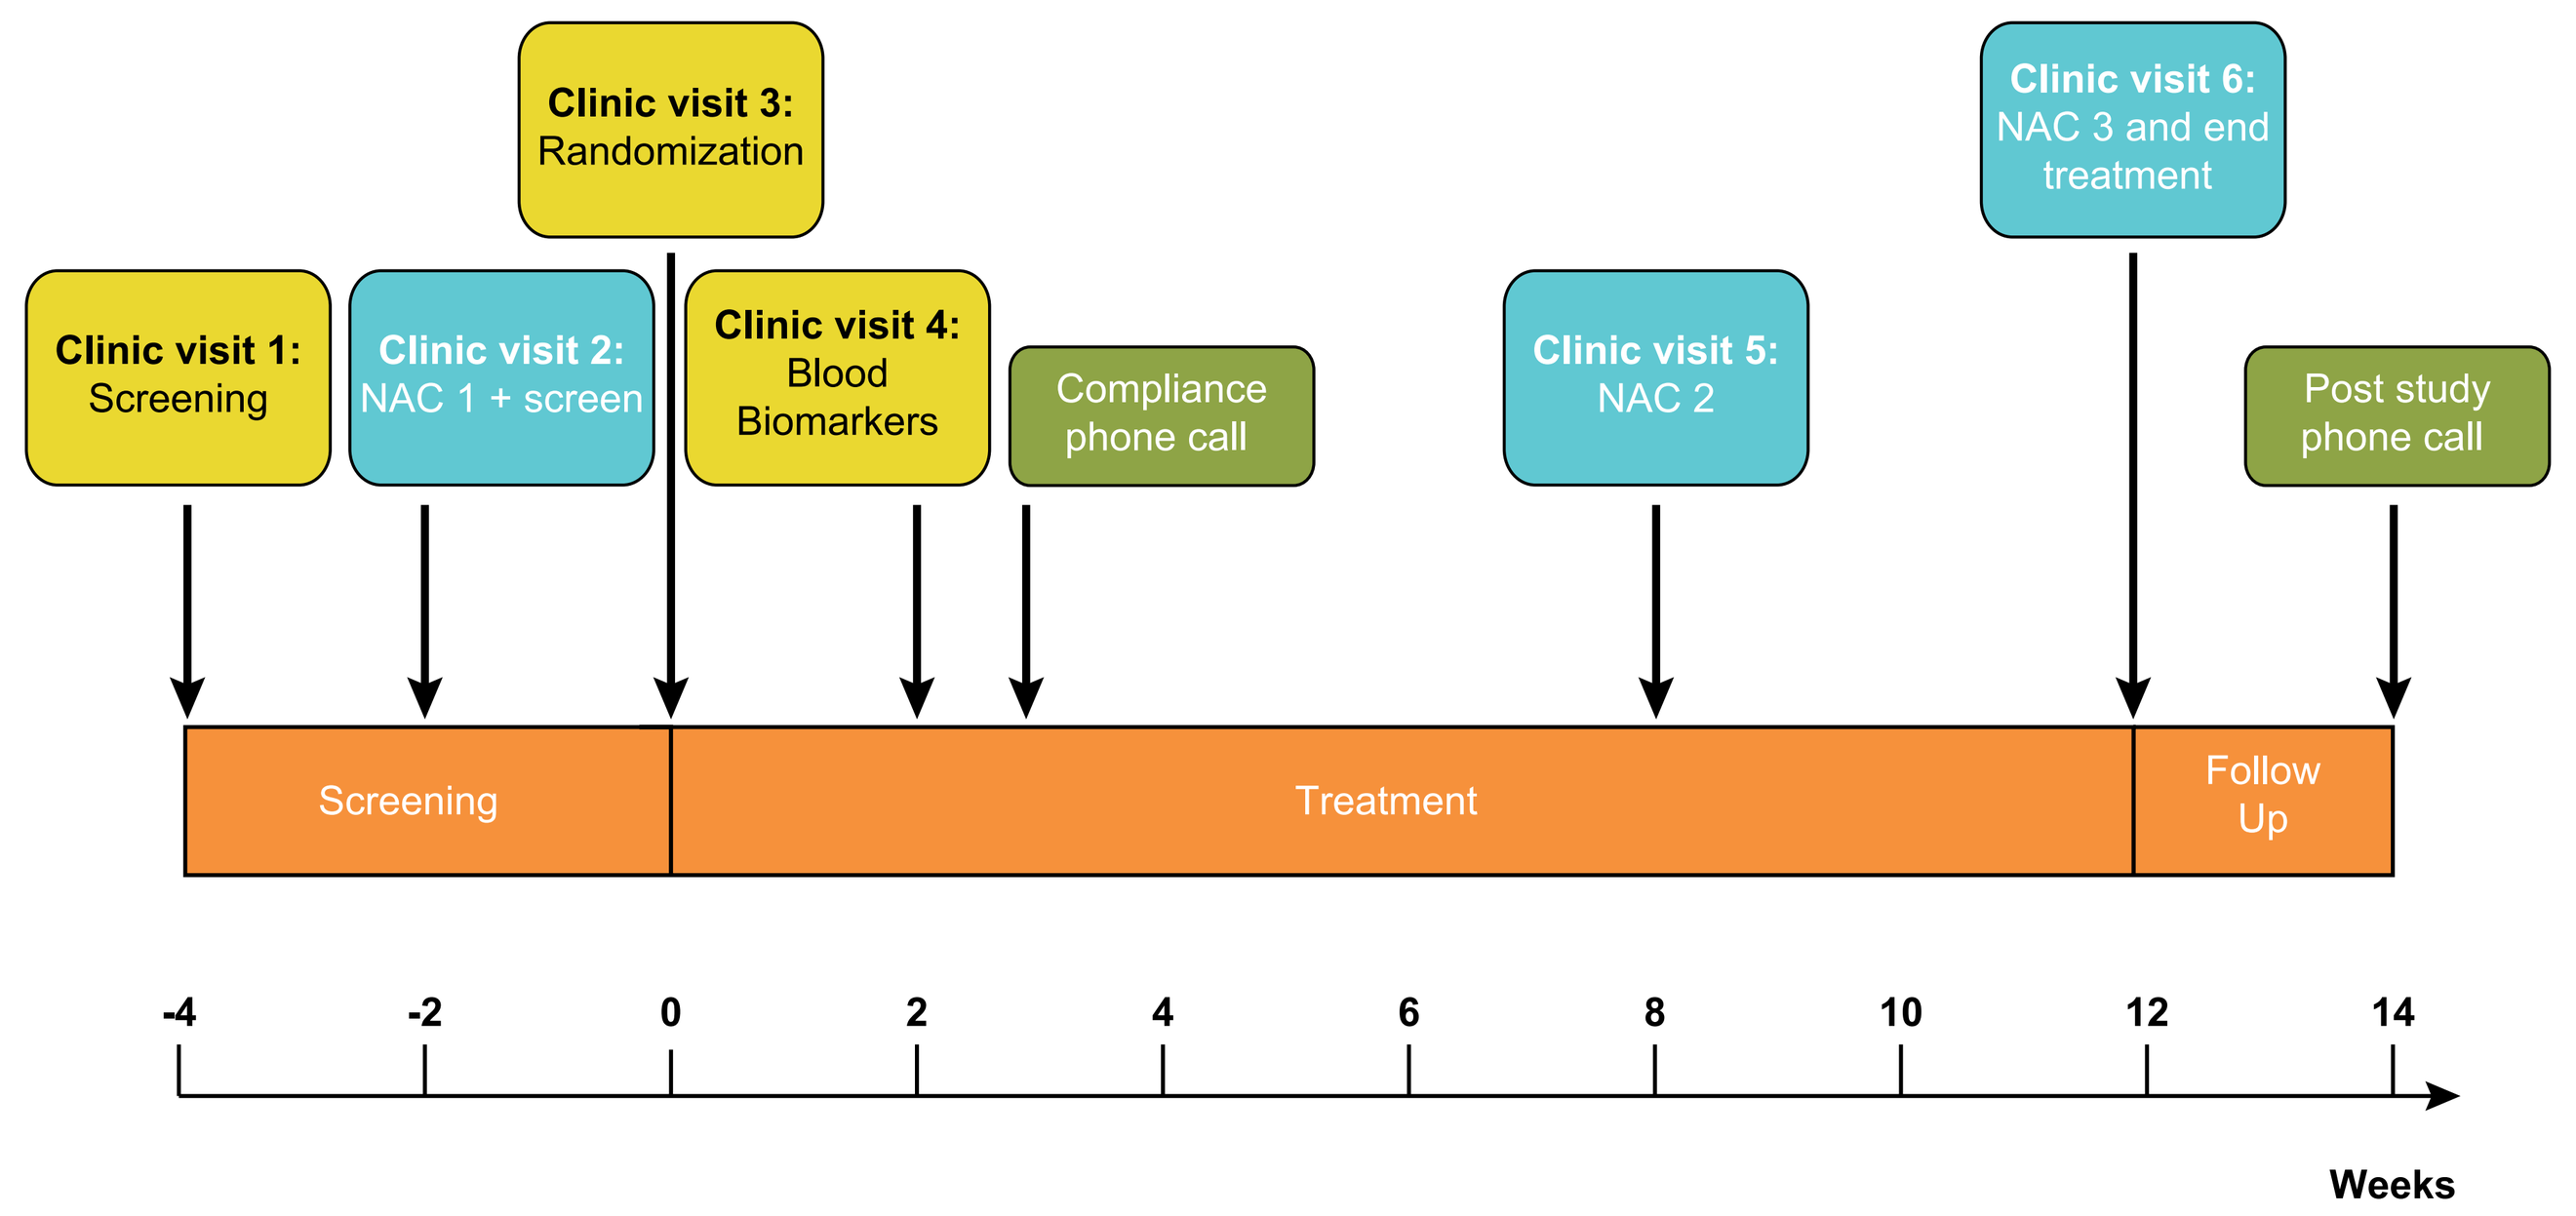
Clinic visit 1: Screening
Clinic visit 2: NAC 1 + screen
Clinic visit 3: Randomization
Clinic visit 4: Blood Biomarkers
Compliance phone call
Clinic visit 5: NAC 2
Clinic visit 6: NAC 3 and end treatment
Post study phone call
Screening
Treatment
Follow Up
-4 -2 0 2 4 6 8 10 12 14
Weeks

B)

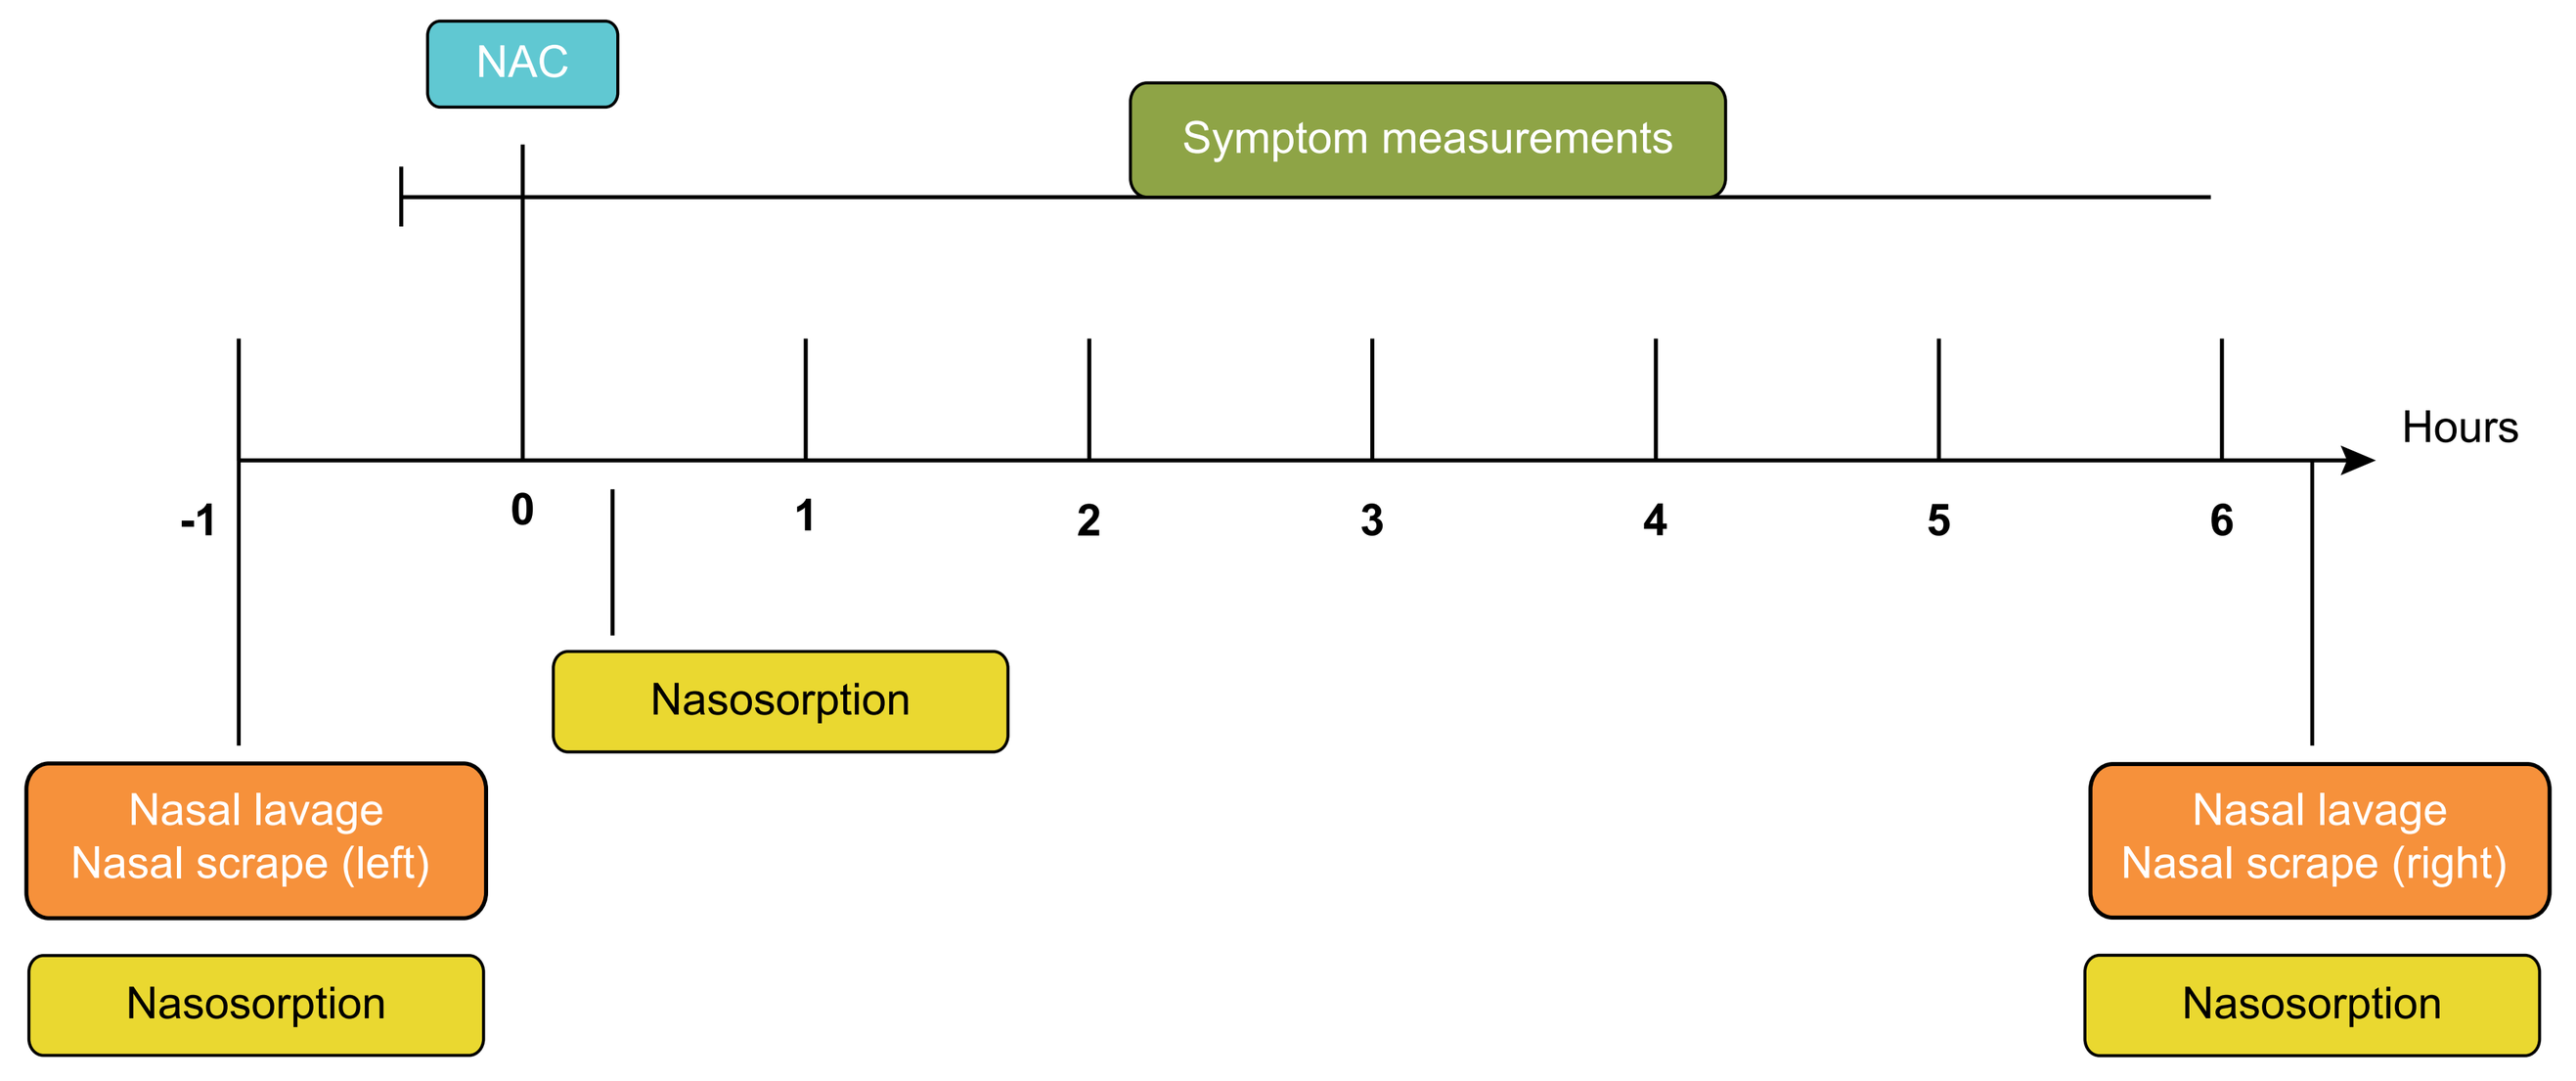
NAC
Symptom measurements
Hours
-1 0 1 2 3 4 5 6
Nasosorption
Nasal lavage Nasal scrape (left)
Nasosorption
Nasal lavage Nasal scrape (right)
Nasosorption

Results

- A total of 23 subjects were randomized (n=16, 12 SQ-HDM; n=7, placebo) and 21 subjects completed the study
- HDM-specific IgG_4 significantly increased from baseline with 12 SQ-HDM versus placebo, increasing at week 8 and increasing further by week 12 (**Figure 2**)
- IgE-BF significantly increased with 12 SQ-HDM versus placebo at both weeks 8 and 12 (**Figure 2**)
- In comparison to baseline NAC, treatment with 12 SQ-HDM reduced early symptoms by 43% at week 8 and 57% at week 12 (**Figure 3**)
- In comparison to baseline NAC, treatment with 12 SQ-HDM reduced peak symptoms (15 minutes post NAC) by 36% at week 8 and 52% at week 12

Results (continued)

Figure 2. IgG_4 and IgE-BF. *P<0.05 for difference in ratio of geometric mean (IgG_4) or change from baseline (IgE-BF) vs placebo determined by constrained longitudinal data analysis (cLDA) model. HDM, house dust mite.

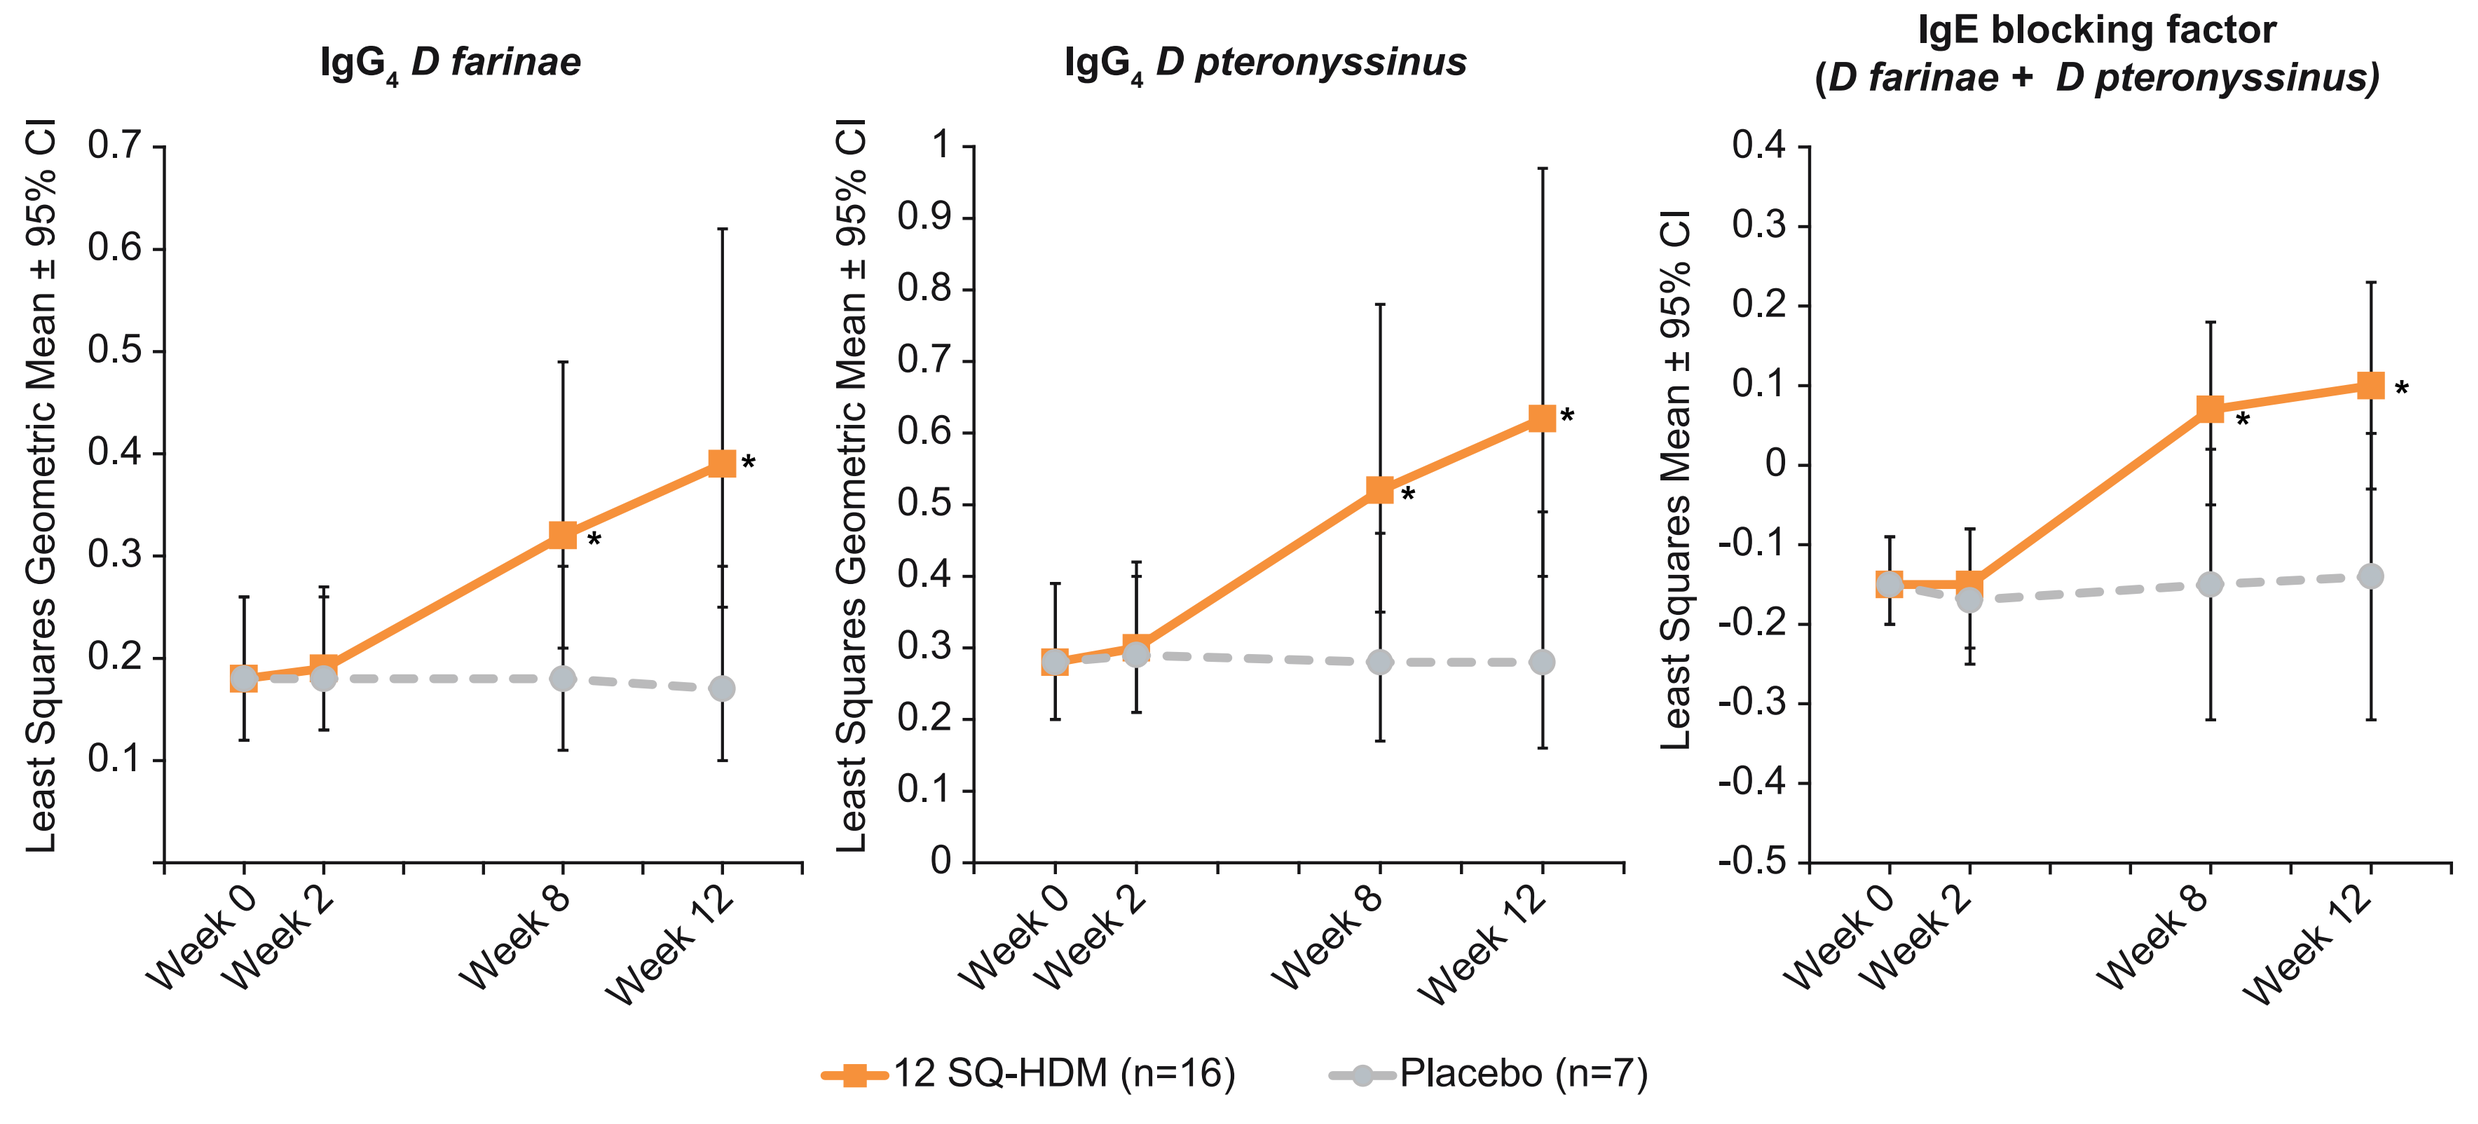
IgG4 D farinae
IgG4 D pteronyssinus
IgE blocking factor (D farinae + D pteronyssinus)
Least Squares Geometric Mean ± 95% CI
Least Squares Mean ± 95% CI
Week 0 Week 2 Week 8 Week 12
12 SQ-HDM (n=16)
Placebo (n=7)

Figure 3. Nasal symptoms. TNSS during A) NAC and B) early and late phases of NAC at weeks −2, 8, and 12. Percentages indicate changes in time-weighted score from baseline. *P≤0.05 for change from baseline vs placebo using cLDA model. HDM, house dust mite; NAC, nasal allergen challenge.

A)

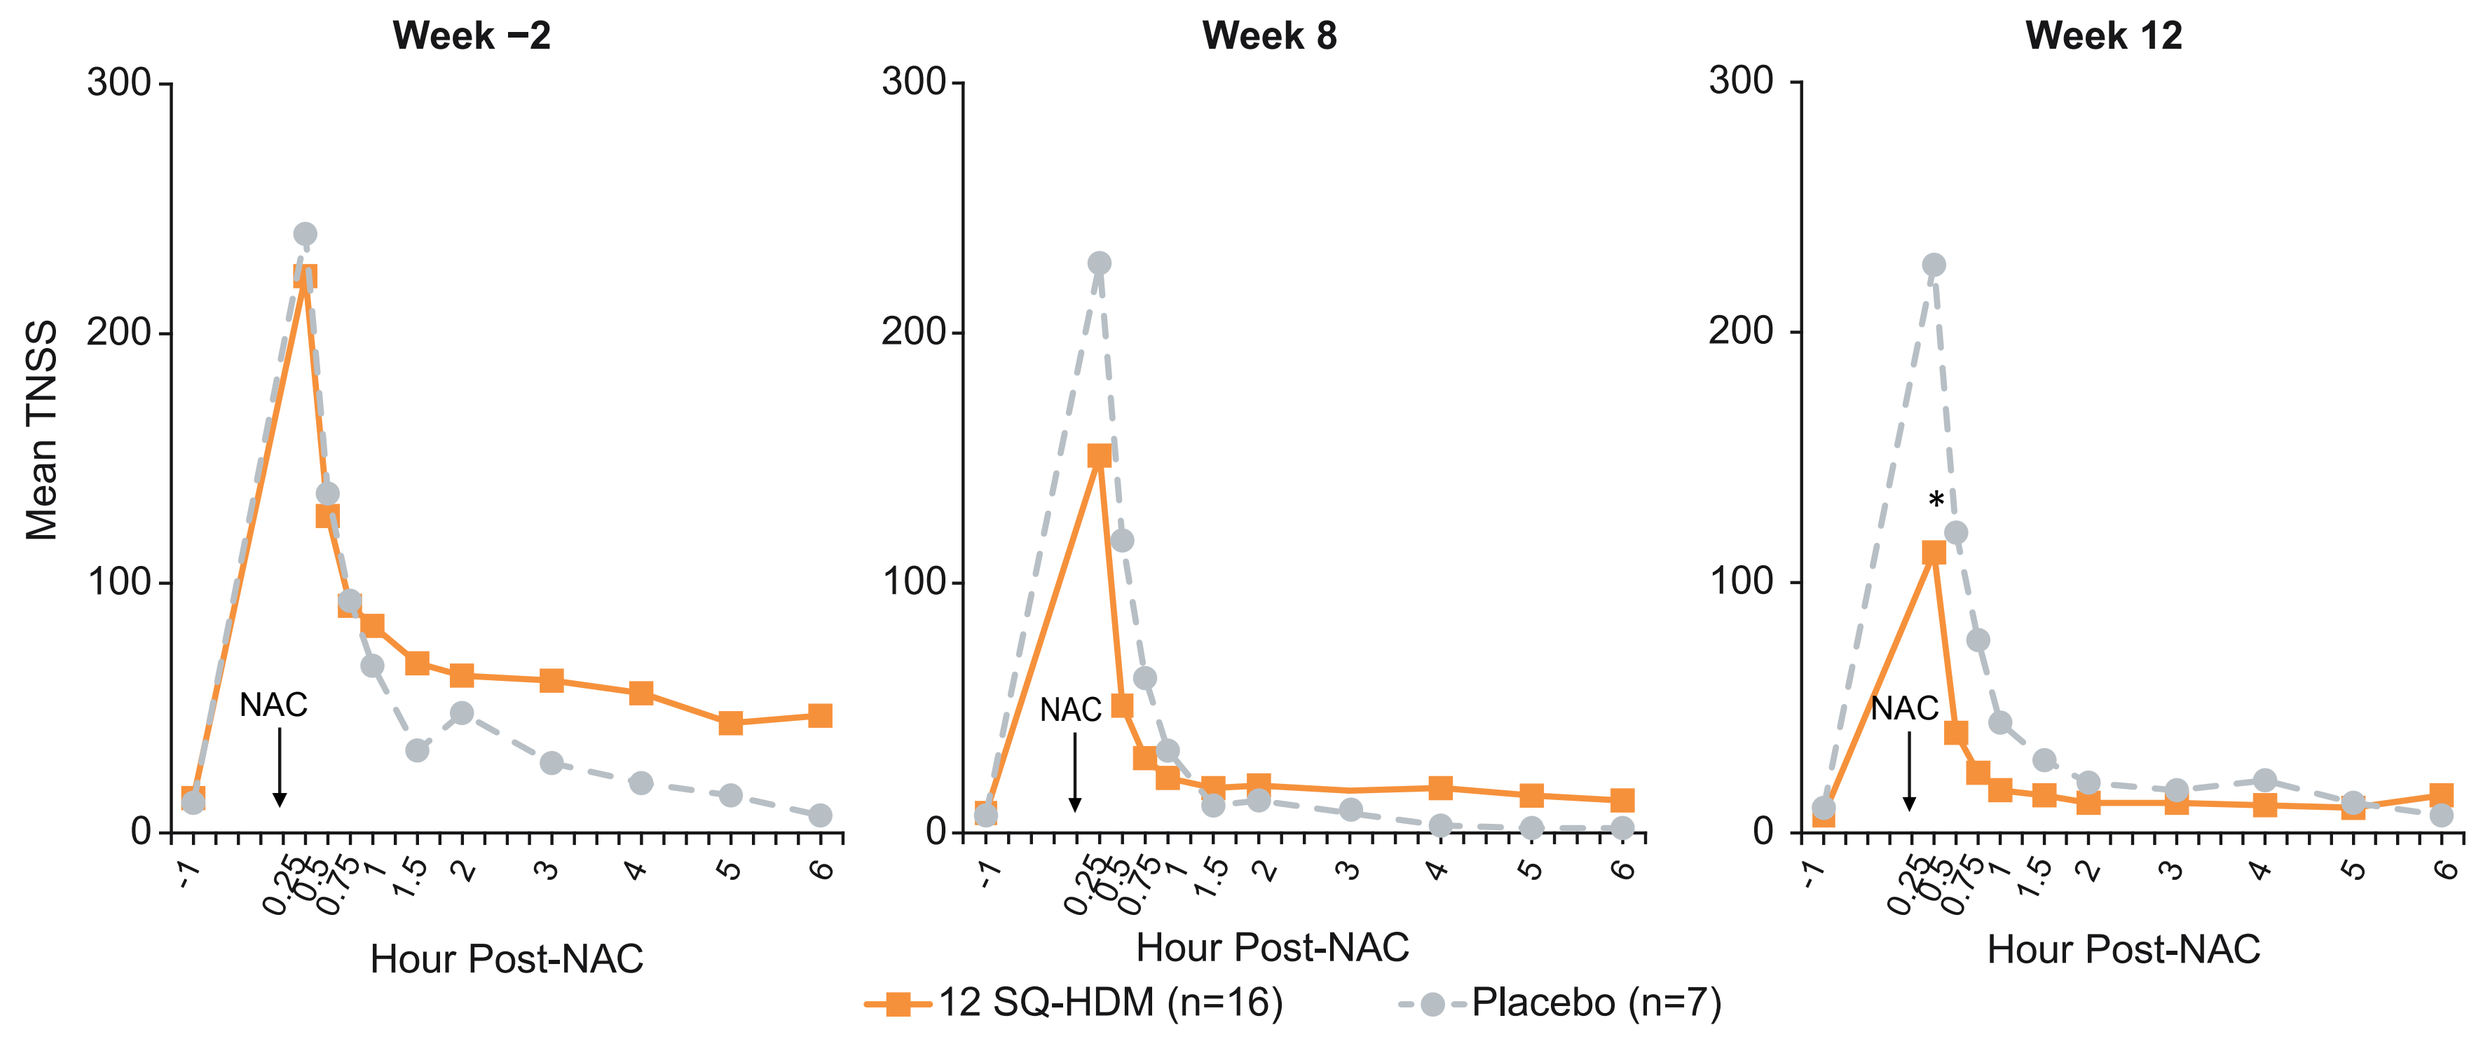
Week −2
Week 8
Week 12
Mean TNSS
NAC
Hour Post-NAC
12 SQ-HDM (n=16)
Placebo (n=7)

B)

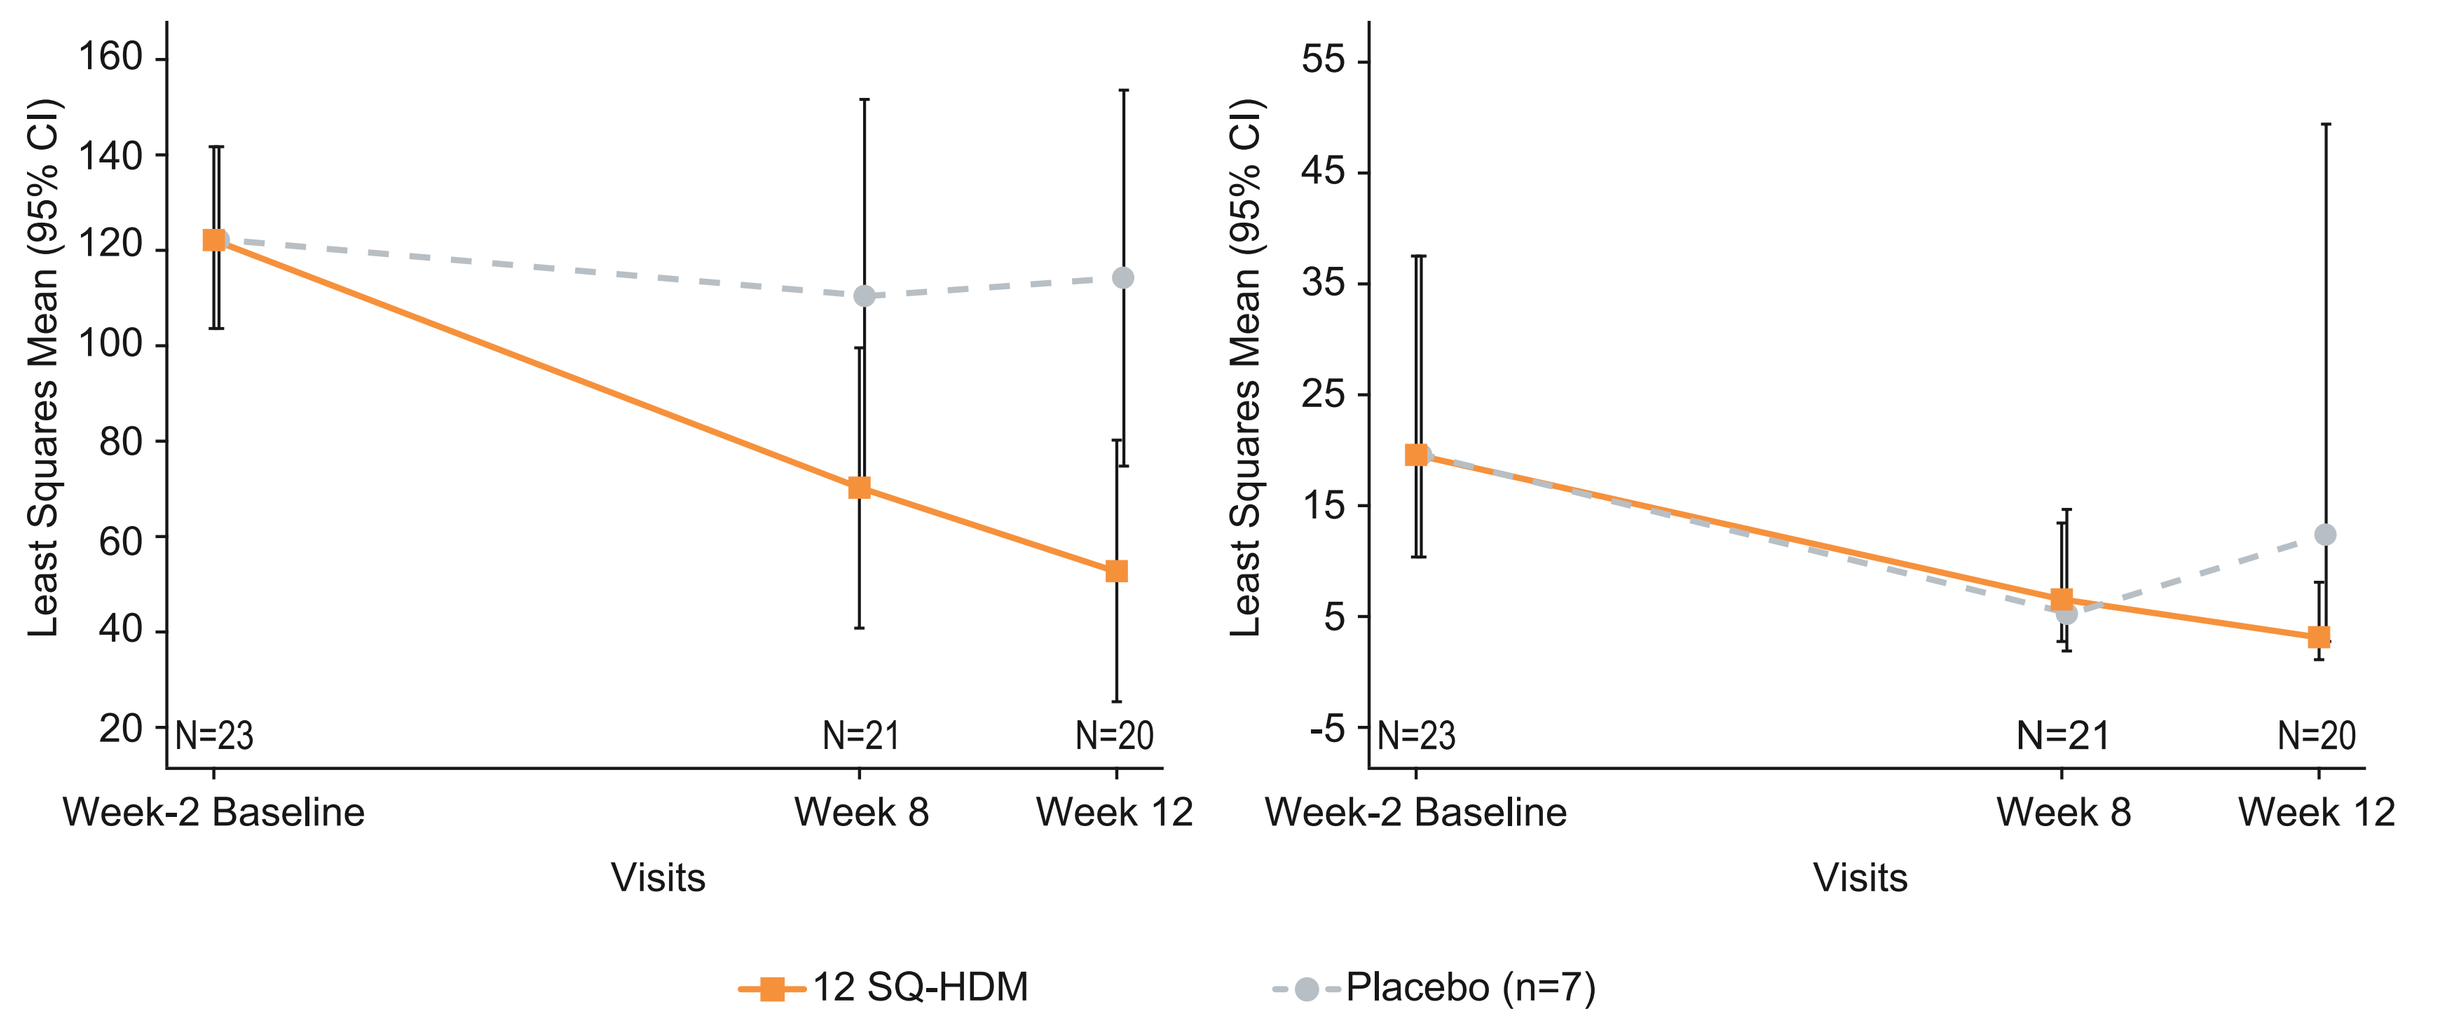
Least Squares Mean (95% CI)
N=23 N=21 N=20
Week-2 Baseline Week 8 Week 12
Visits
12 SQ-HDM
Placebo (n=7)

- There was no significant difference between the 12 SQ-HDM group and placebo in change from baseline for PNIF during any NAC challenge
- No significant differences from baseline NAC or in fold changes from −1 hour pre NAC to 6.5 hours post NAC between 12 SQ-HDM and placebo were observed for IL-5 at any timepoint (**Figure 4**)
 - There were also no significant differences for IL-13 or TARC

Figure 4. Nasal concentrations of IL-5 in mucosal lining fluid at weeks −2, 8, and 12 NAC. Box plots indicate medians and quartiles. Error bars indicate minimum and maximum values. Fold changes are estimated and the fold change at baseline is assumed to be the same for both treatment groups. *P* values were determined using the cLDA model. HDM, house dust mite; NAC, nasal allergen challenge.

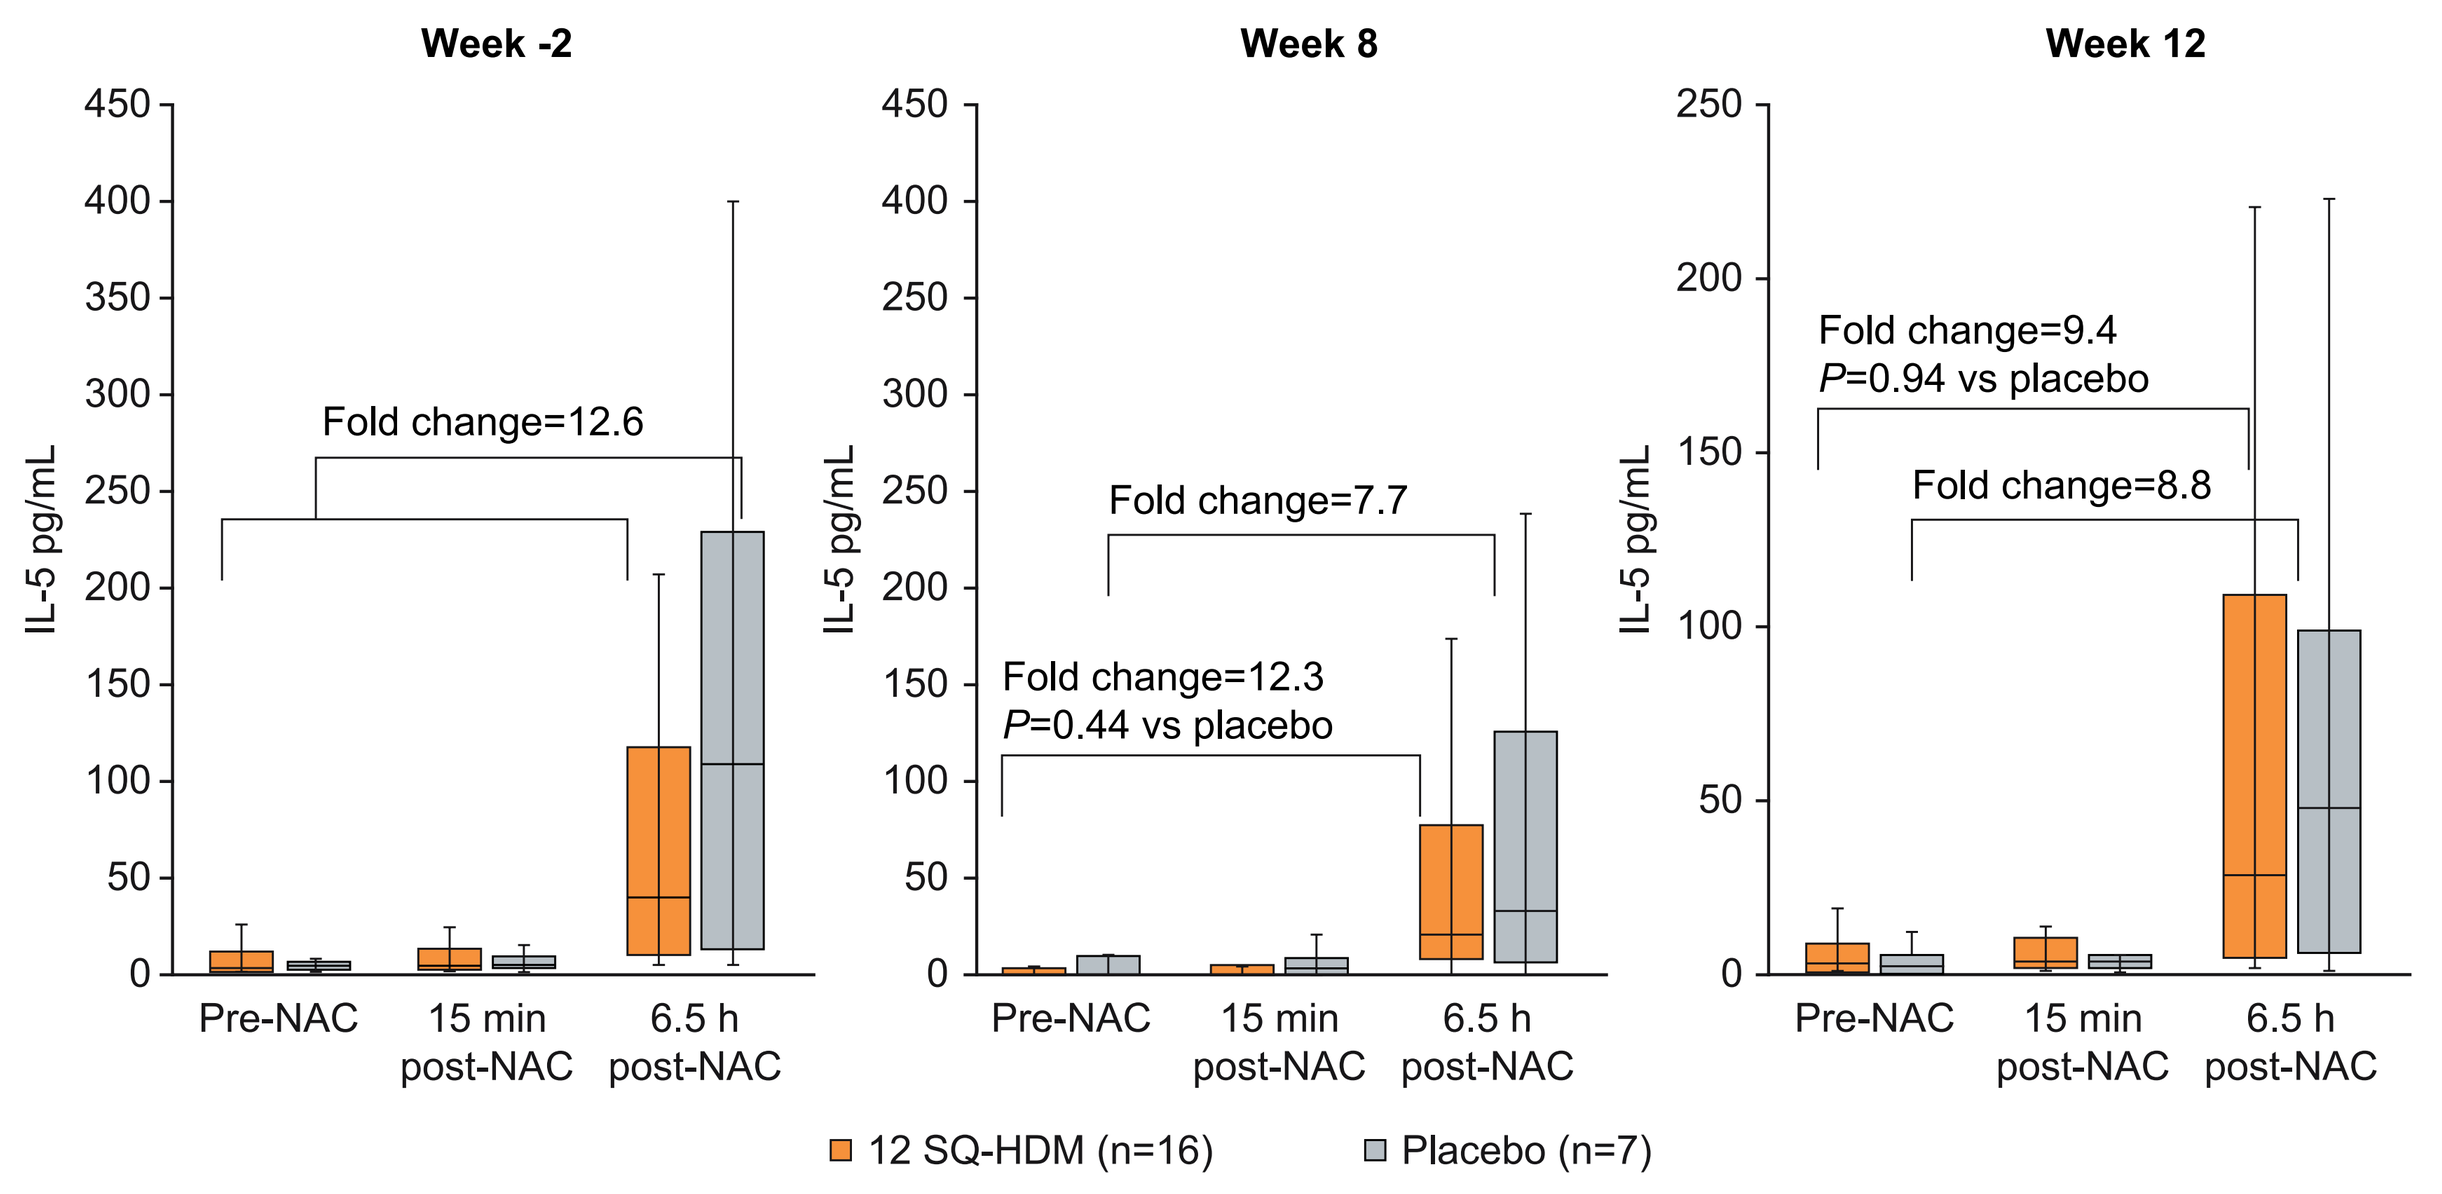
Week -2
Week 8
Week 12
IL-5 pg/mL
Fold change=12.6
Fold change=7.7
Fold change=12.3 P=0.44 vs placebo
Fold change=9.4 P=0.94 vs placebo
Fold change=8.8
Pre-NAC
15 min post-NAC
6.5 h post-NAC
12 SQ-HDM (n=16)
Placebo (n=7)

- There was a good correlation between absolute values for known mRNA markers of mast cells, IL-5 and IL-13, and eosinophils with most r values ≥0.90
- A trend toward reduction in eosinophil, mast cell, and Th2 inflammation markers from baseline in 12-SQ HDM treated group versus placebo was observed, but did not reach statistical significance (**Figure 5**)
 - There were no consistent correlations found between these markers and VAS symptoms across all the NAC timepoints, either with placebo or 12 SQ-HDM

Figure 5. Change from baseline NAC in eosinophil, mast cell, or Th2 inflammation mRNA markers before and after week 8 and week 12 NAC, and fold change from baseline NAC.

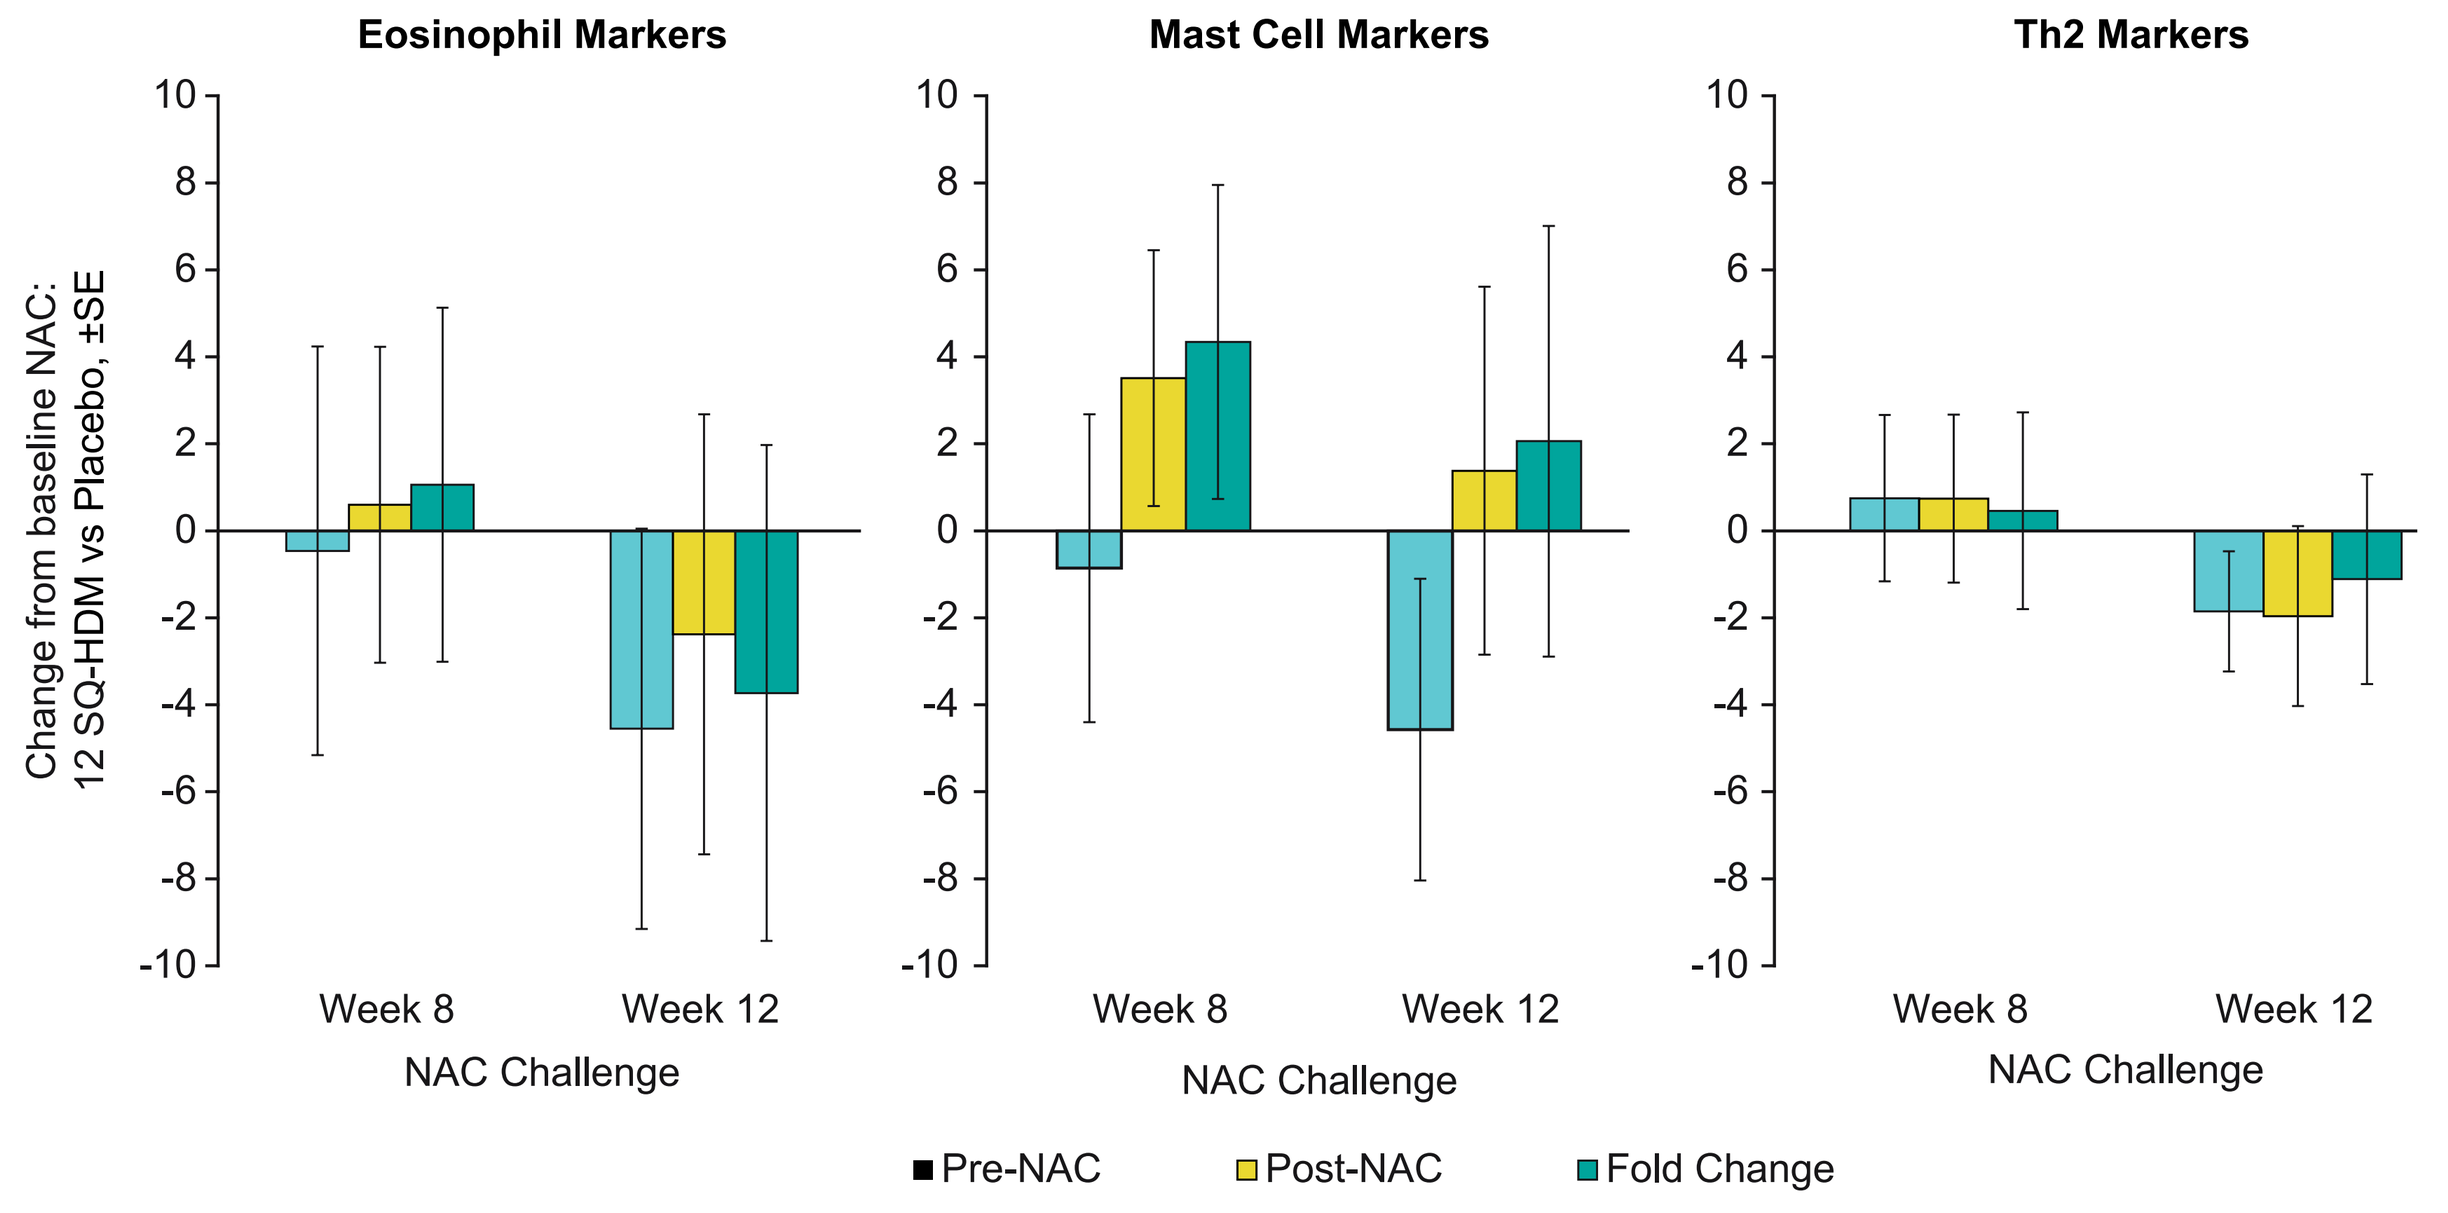
Eosinophil Markers
Mast Cell Markers
Th2 Markers
Change from baseline NAC: 12 SQ-HDM vs Placebo, ±SE
Week 8 Week 12
NAC Challenge
Pre-NAC
Post-NAC
Fold Change

- No serious AEs, systemic allergic events, or epinephrine administrations were reported
- The most frequent AEs were upper respiratory tract infection, throat irritation, tongue pruritus, mouth swelling, and oral paresthesia
- Two events of mild local hypersensitivity reactions were reported

Conclusions

- Induction of HDM-specific IgG_4 and IgE-BF by 12 SQ-HDM, along with significant improvement in early phase NAC-induced nasal symptoms, suggests that IgE-BF generation may contribute to the mechanism of action of 12 SQ-HDM during the first 12 weeks of treatment primarily affecting the early allergic response
- In this study there was no significant effect on mucosal IL-5 and IL-13, or eosinophil-associated gene expression demonstrated
- Treatment was well tolerated and the AE profile was consistent with that reported in large clinical trials of 12 SQ-HDM[1,2]

Acknowledgments

The authors would like to thank Ashish Jain and Laura George for their contributions to the study. Tim Mant is also supported by the National Institute for Health Research (NIHR) Biomedical Research Centre at Guy's and St Thomas' NHS Foundation Trust and King's College London. Medical writing and editorial assistance were provided by Erin P. Scott, PhD, of Scott Medical Communications, LLC. This assistance was funded by Merck & Co., Inc., Kenilworth, NJ, USA.

Disclosures

Q. Zhao, K. Tsai, D. Selverian, L.N. Carayannopoulos, and H. Nolte are employees of Merck Sharp & Dohme Corp., a subsidiary of Merck & Co., Inc., Kenilworth, NJ, USA. K. Lund is an employee of ALK. N.C. Gunawardana has nothing to disclose. T.T. Hansel is establishing a company called Mucosal Diagnostics. G.W. Clarke was an employee of Quintiles IMS, Reading, UK, and T. Mant is a current employee of Quintiles IMS, which provides consulting services for Merck Sharp & Dohme Corp.

Funding

Funding for this research was provided by Merck & Co., Inc., Kenilworth, NJ, USA.

References

1. Virchow JC, Backer V, Kuna P, et al. Efficacy of a house dust mite sublingual allergen immunotherapy tablet in adults with allergic asthma: a randomized clinical trial. *JAMA*. 2016;315(16):1715-1725.
2. Nolte H, Bernstein DI, Nelson HS, et al. Efficacy of house dust mite SLIT-tablet in North American adolescents and adults in a randomized, placebo-controlled trial. *J Allergy Clin Immunol*. 2016;138(6):1631-1638.

*Current affiliation in hVIVO Ltd, London, UK

Copyright © 2017 Merck Sharp & Dohme Corp., a subsidiary of **Merck & Co., Inc.** All rights reserved.